A mechanistic computational model of the activation of platelets by ADP, collagen, TxA_2 and thrombin and its inhibition by ASA and thienopyridines

Rolf Burghaus[1], Gertrud Ahr[2], Katrin Coböken[1], Evin Hizaler[3], Lars Kuepfer[3], Jörg Lippert[3], Christoph Niederalt[3], Elisabeth Perzborn[4], Jochen Schaub[3], Anke Sensse[3], Hans-Ulrich Siegmund[3]

Bayer Technology Services

1) Computational Solutions, Bayer Technology Services GmbH, Leverkusen, Germany

2) Clinical Pharmacology, Bayer HealthCare AG, Wuppertal, Germany

3) Systems Biology, Bayer Technology Services GmbH, Leverkusen, Germany

4) PRR Pharmacology, Bayer HealthCare AG, Wuppertal, Germany

Bayer HealthCare

INTRODUCTION

- Platelets play a fundamental role in blood coagulation. Their physiological functions include provision of binding sites for coagulation factors, excretion of procoagulant factors, and mechanical closure of vessel rupture.
- **OBJECTIVE:** To better understand the impact of platelet activation in the coagulation process for both the *in vitro* [platelet rich plasma (e.g. Hemker's ETP test in PRP)] and *in vivo* scenarios, we developed a computational model representing platelet activation and their inhibition by cyclooxygenase blockers [aspirin (ASA)] and ADP receptor blockers (thienopyridines).
- The model differentiates between weak (ADP) and strong activating agents (TxA2, collagen, thrombin) and directly represents induction of thromboxane synthesis, ADP and factor V secretion, membrane flip-flop, as well as activation of P2Y1, P2Y12, PAR1 and PAR4.
- We coupled the platelet activation model to a model of the biochemical coagulation cascade. First we attributed published numbers of factor binding sites to non-activated and activated platelets, and secondly we increased activation kinetics at platelets compared to reactions in plasma [3].

MODELS

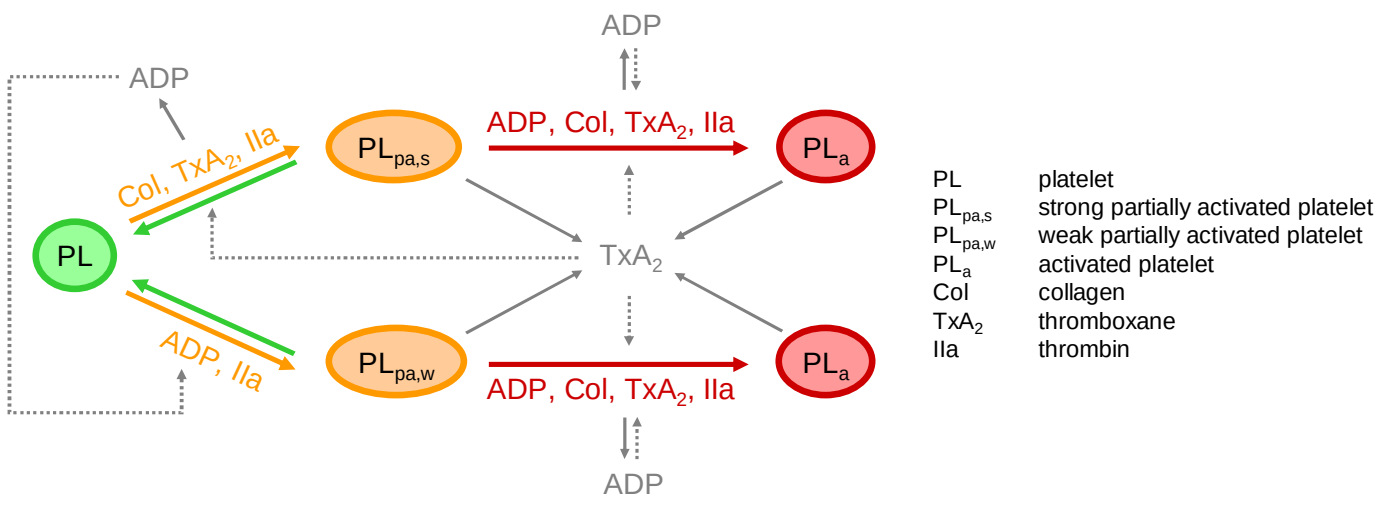
ADP
ADP
Col, TxA2, IIa
PL
PLpa,s
PLpa,w
ADP, Col, TxA2, IIa
PLa
TxA2
ADP, IIa
ADP, Col, TxA2, IIa
PLa
ADP
PL platelet
PLpa,s strong partially activated platelet
PLpa,w weak partially activated platelet
PLa activated platelet
Col collagen
TxA2 thromboxane
IIa thrombin

Figure 1: Sketch of platelet activation model. The model differentiates between a weak (ADP, IIa via PAR1) and a strong (Col, TxA_2, IIa via PAR4) path of platelet activation.

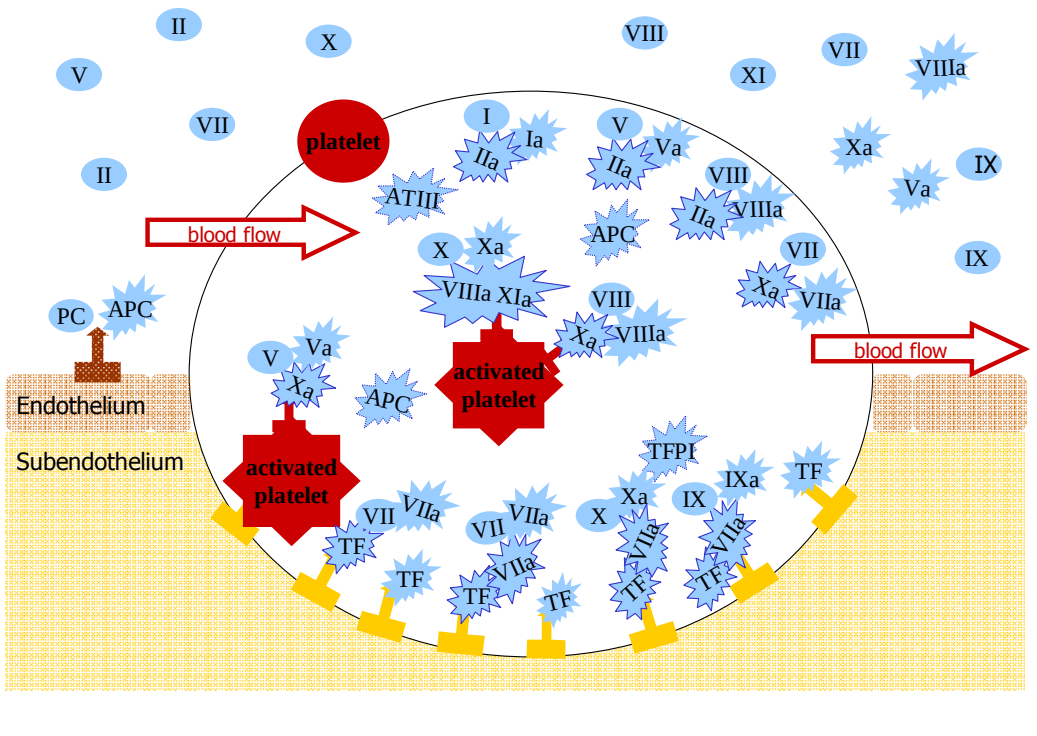
platelet
blood flow
blood flow
activated platelet
activated platelet
Endothelium
Subendothelium
PC
APC
TFPI

Figure 2: Scheme of the blood coagulation model including the coagulation factor cascade, the platelet activation and the compartmentalization of the reaction space. Factor and platelet activation take place on the sub-endothelium, on the platelet surface or in plasma. Protein C is activated on the endothelium.

MoBi IMPLEMENTATION

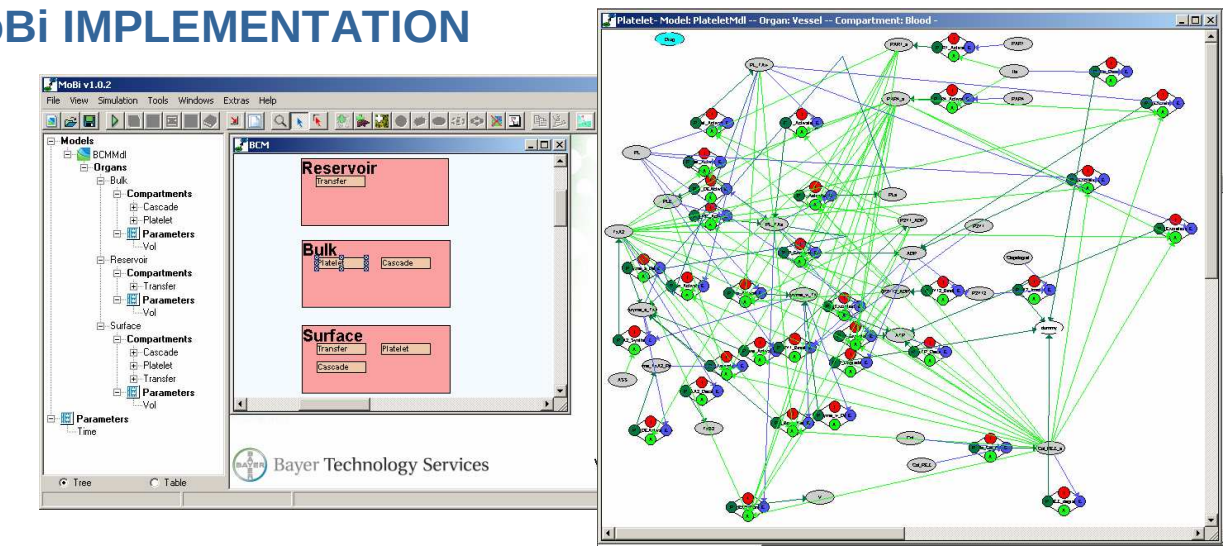
MoBi v1.0.2
Reservoir
Bulk
Surface
Bayer Technology Services
Platelet- Model: PlateletMdl -- Organ: Vessel -- Compartment: Blood -

CONCLUSIONS

- The core platelet model accurately describes a broad spectrum of *in vitro* studies with citrate buffered plasma including combination treatments with ADP plus either collagen or thrombin with and without ASA or thienopyridine [1,2].
- The model explains both the dynamics of ADP excretion and transmission experiments where light transmission increases due to platelet aggregation.
- The integrated model of platelets interacting with the coagulation cascade is in accordance with studies of Hemker's ETP assay with PRP [4,5].

RESULTS

Platelet activation: Transmission and ADP secretion

Platelet model

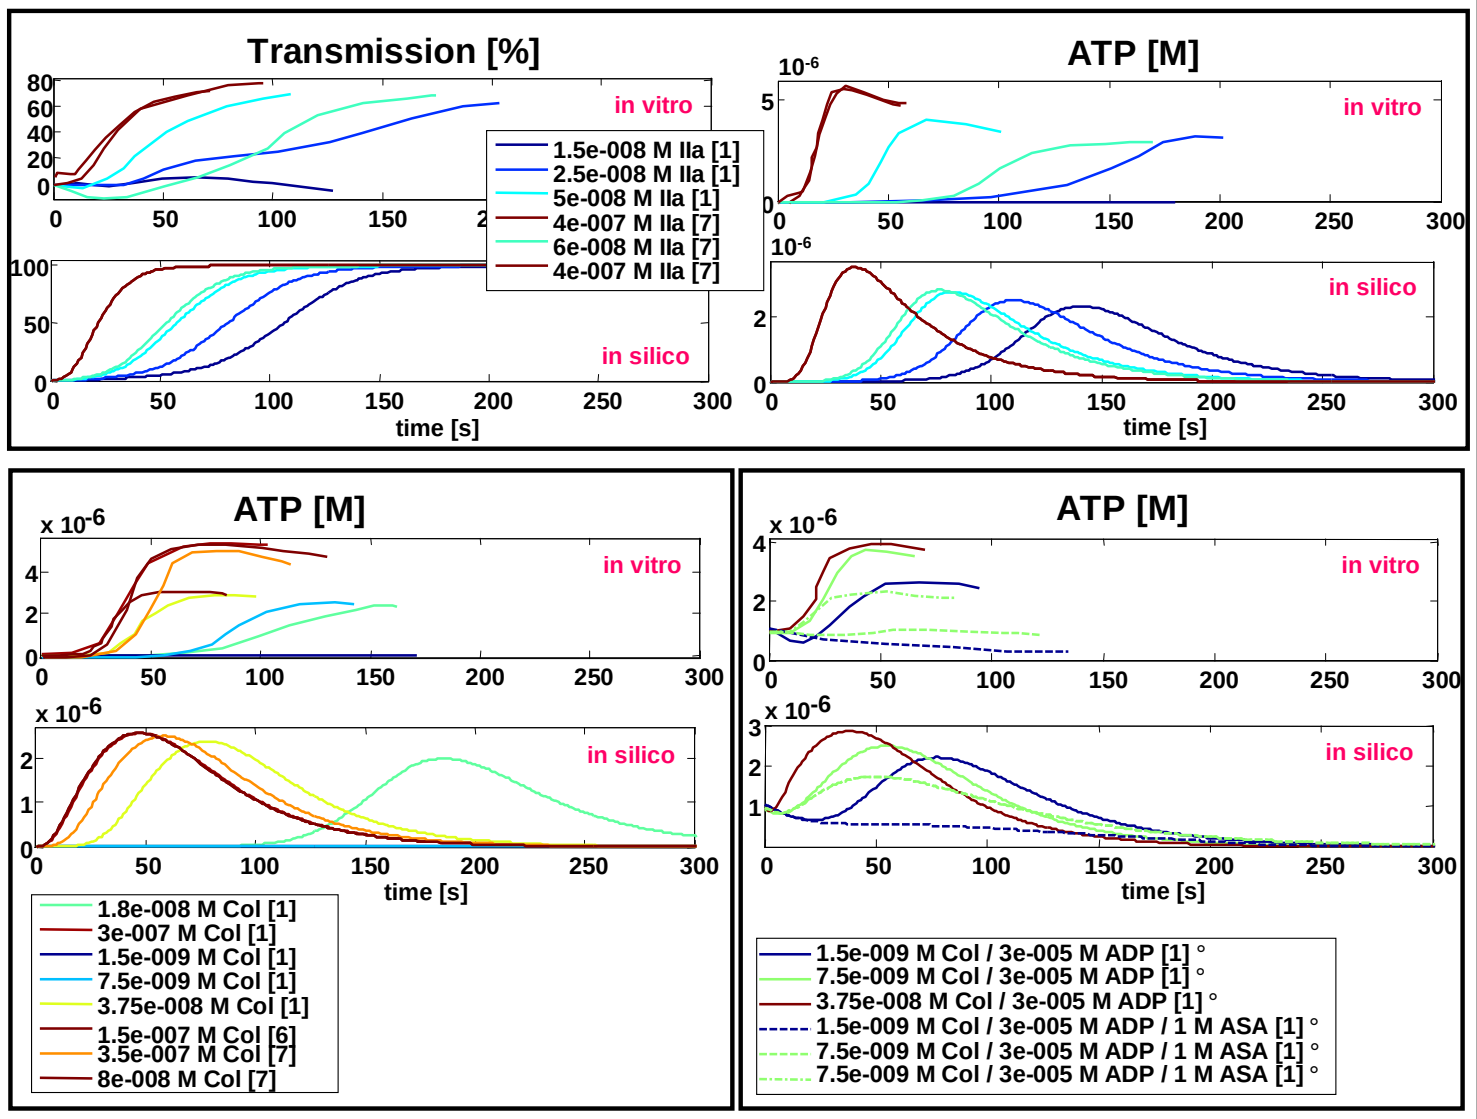
Transmission [%]
ATP [M]
in vitro
in silico
time [s]
1.5e-008 M IIa [1]
2.5e-008 M IIa [1]
5e-008 M IIa [1]
4e-007 M IIa [7]
6e-008 M IIa [7]
4e-007 M IIa [7]
1.8e-008 M Col [1]
3e-007 M Col [1]
1.5e-009 M Col [1]
7.5e-009 M Col [1]
3.75e-008 M Col [1]
1.5e-007 M Col [6]
3.5e-007 M Col [7]
8e-008 M Col [7]
1.5e-009 M Col / 3e-005 M ADP [1] °
7.5e-009 M Col / 3e-005 M ADP [1] °
3.75e-008 M Col / 3e-005 M ADP [1] °
1.5e-009 M Col / 3e-005 M ADP / 1 M ASA [1] °
7.5e-009 M Col / 3e-005 M ADP / 1 M ASA [1] °
7.5e-009 M Col / 3e-005 M ADP / 1 M ASA [1] °

Figure 3: Platelet aggregation (light transmission) and ADP secretion induced by either single or combinatorial triggering with collagen (Col), ADP and thrombin (IIa). The upper and lower rows present the original experimental results and the simulation results obtained with the platelet model, respectively. Additionally, the effect of Aspirin (ASA) on TxA_2 formation is analysed. Samples marked with ° contained ATP impurities.

Integrated model

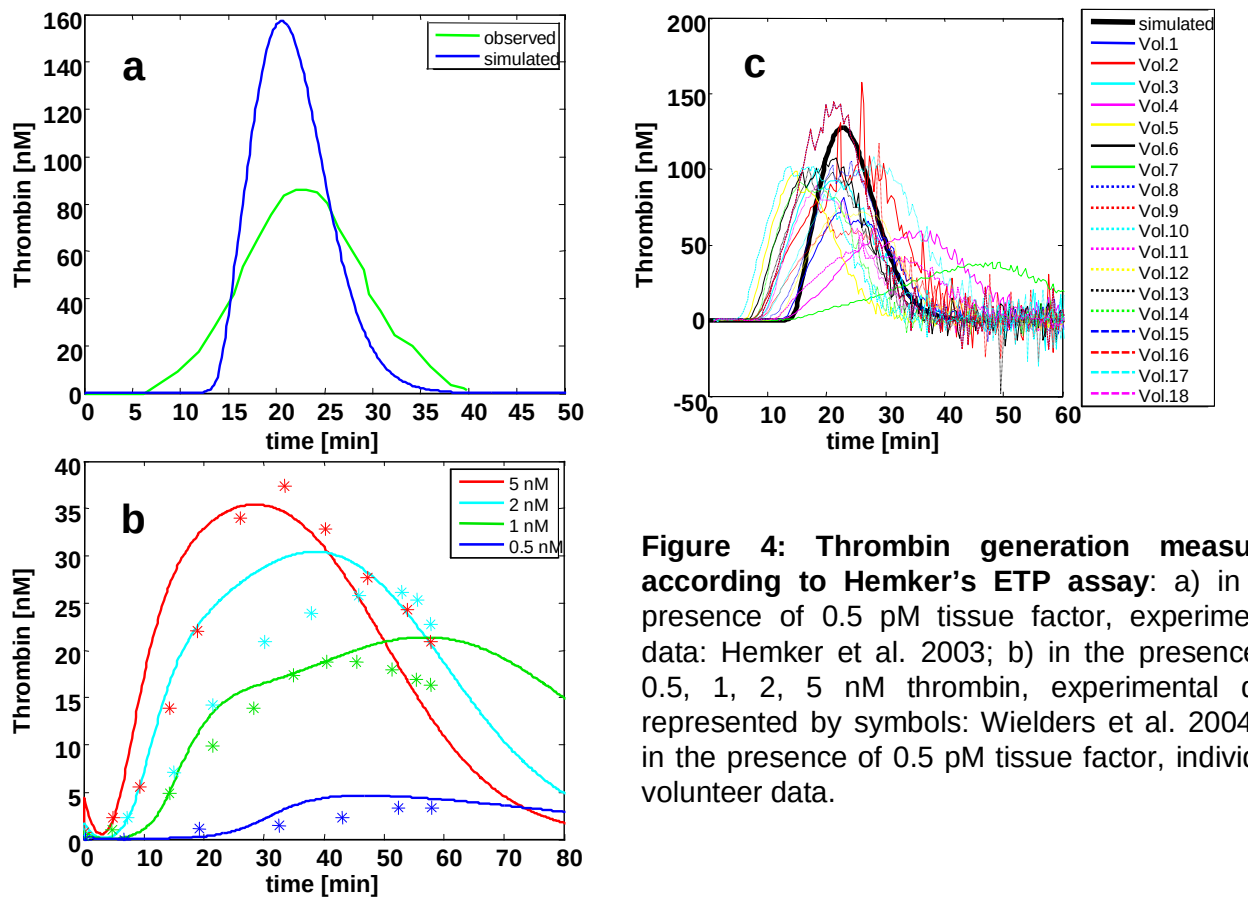
a
b
c
Thrombin [nM]
time [min]
observed
simulated
5 nM
2 nM
1 nM
0.5 nM
simulated
Vol.1
Vol.2
Vol.3
Vol.4
Vol.5
Vol.6
Vol.7
Vol.8
Vol.9
Vol.10
Vol.11
Vol.12
Vol.13
Vol.14
Vol.15
Vol.16
Vol.17
Vol.18

Figure 4: Thrombin generation measured according to Hemker's ETP assay: a) in the presence of 0.5 pM tissue factor, experimental data: Hemker et al. 2003; b) in the presence of 0.5, 1, 2, 5 nM thrombin, experimental data represented by symbols: Wielders et al. 2004; c) in the presence of 0.5 pM tissue factor, individual volunteer data.

References: [1] Huang EM, Detwiler TC. Blood 1981;57:685-91. [2] Savi P et al. Thromb Haemost 2000;84:891-6. [3] Colman RW et al. Hemostasis and Thrombosis: Basic Principles and Clinical Practice 5th Ed. 2006. Lippincott Williams & Wilkins, Philadelphia, PA. [4] Hemker HC et al. Pathophysiol Haemost Thromb 2003;33:4-15. [5] Wielders SJH et al. Arterioscler Thromb Vasc Biol 2004;24:1138-1142. [6] De Caterina R et al. J Nucl Med Allied Sci 1984; 28(3):185-96. [7] Charo IF et al. Nature 1977; 269(5623):66-9.