Computational investigation of optimal dosing schedules for a switch of medication from Warfarin to direct inhibitors of vitamin K dependent factors

Rolf Burghaus[1], Gertrud Ahr[2], Katrin Coböken[1], Jörg Lippert[3], Frank Misselwitz[4], Wolfgang Mück[2], Christoph Niederalt[3], Anke Sensse[3], Hans-Ulrich Siegmund[3]

Bayer Technology Services

1) Computational Solutions, Bayer Technology Services GmbH, Leverkusen, Germany
2) Clinical Pharmacology, Bayer Healthcare AG, Wuppertal, Germany
3) Systems Biology, Bayer Technology Services GmbH, Leverkusen, Germany
4) Global Clinical Development CV, Bayer Healthcare AG, Wuppertal, Germany

Bayer HealthCare

INTRODUCTION

- Warfarin therapy results in a significant reduction of the concentrations of vitamin K dependent coagulation factors II, VII, IX, and X. After discontinuation of Warfarin application, normal coagulation behavior recovers only after several days due to slow factor turn-around times.
- If the medication is switched from Warfarin to another drug, like an inhibitor of activated factor II or X, this slow recovery behavior must be accounted for by dose adjustments for the prevention of increased bleeding during the period of medication switch.
- **OBJECTIVE:** To study the joint effect of Warfarin and inhibitors of coagulation factors like Ximelagatran, DX-9065a or Enoxaparin by means of a computational model. The model mechanistically represents the interaction of different classes of drugs. We derive time-dependent dosing windows for the newly started therapy considering efficacy as well as safety.

METHODS

Exploitation of an integrated computational model consisting of

- the suppression of vitamin K dependent factors due to Warfarin (**Fig. 1**) and
- the coagulation cascade

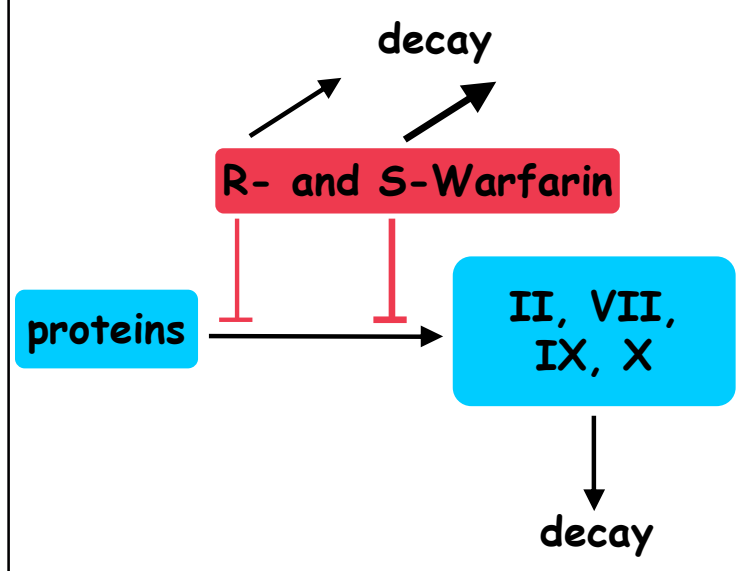
decay
R- and S-Warfarin
proteins
II, VII, IX, X
decay

Figure 1. Model of the suppression of vitamin K dependent factors due to Warfarin.

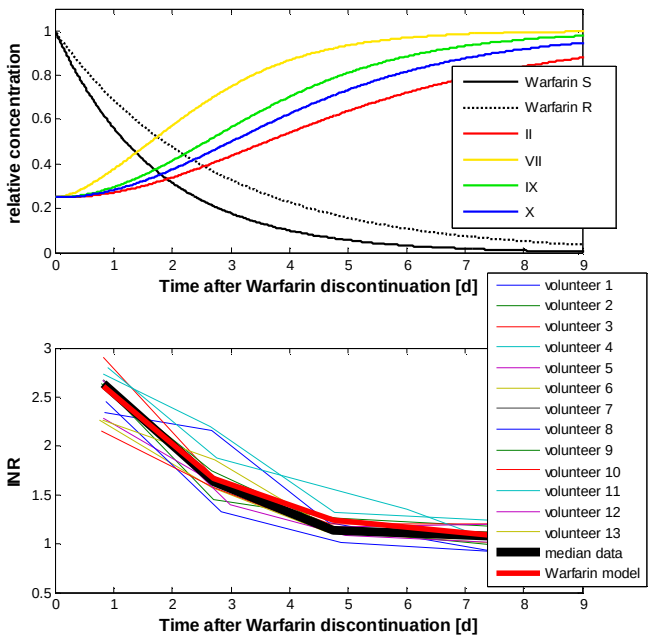
relative concentration
Time after Warfarin discontinuation [d]
Warfarin S
Warfarin R
II
VII
IX
X
INR
volunteer 1
volunteer 2
volunteer 3
volunteer 4
volunteer 5
volunteer 6
volunteer 7
volunteer 8
volunteer 9
volunteer 10
volunteer 11
volunteer 12
volunteer 13
median data
Warfarin model
Time after Warfarin discontinuation [d]

Figure 2. Decay of Warfarin and it's effects. The upper plot presents the simulated Warfarin washout and the recovery of the blood coagulation factors. The lower plot presents INR (International Normalized Ratio) data of 13 volunteers after discontinuation of a Warfarin therapy (provided by R. White, UC Davis) as compared to predictions.

Simulation of the blood coagulation cascade

- Several *in silico* models have been previously described and were considered in this work. The final model is an extension and fusion of models by Hockin et al. (2003), Kogan et al. (2001), Bungay et al. (2003), Orfeo et al. (2004)
- Coagulation factor reaction kinetics and their *in vivo* concentrations were taken from the literature to build up a kinetic model of coupled reactions.
- Validation of this model was done using published experimental results (e.g., PT, aPTT) and internal BHC preclinical and clinical data sets.
- Antithrombotic agents and their actions (DX-9065a, Ximelagatran, Enoxaparin) were added to the kinetic model.
- Efficacy versus safety evaluation was done using different trigger scenarios, some of which were out of any experiment's scope.

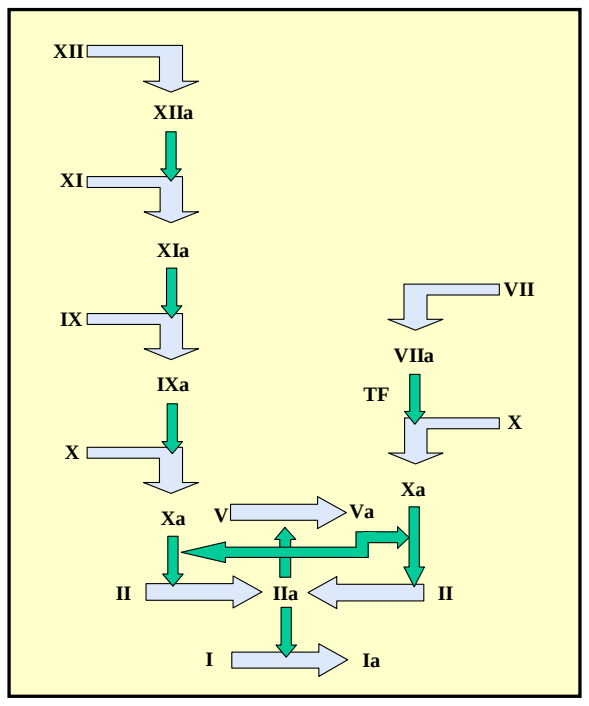
XII
XIIa
XI
XIa
IX
IXa
X
Xa
VII
VIIa
TF
X
Xa
V
Va
II
IIa
II
I
Ia

Figure 2. Blood coagulation cascade. Representation of the backbone of the coagulation process. The complete model considers approximately 70 reactions.

CONCLUSIONS

- Drug-drug interactions can lead to adverse events and are of high relevance both for clinical development and practice. Using a *Systems Biology* approach, we analyzed interactions occurring when Warfarin therapy is discontinued and replaced by inhibitors of the coagulation cascade.
- We predicted clotting properties (like prolongation of the prothrombin time) during the phase of transition. We evaluated the prolongation of clotting caused by combination effects of the different drugs. Our study showed the highly synergistic nature of these interactions and we were able to derive dosing recommendations based on information about individual patients' clearances and disease backgrounds.
- This study is an elegant example of the use of mechanistic modeling approaches for clinical application demonstrating its usefulness for questions of translational medicine and individualized therapy.

RESULTS

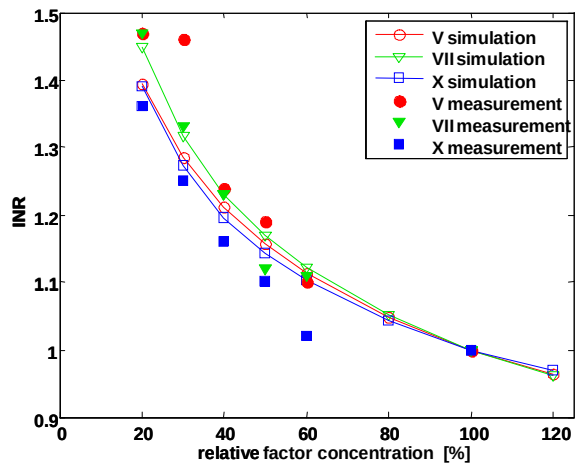
INR
relative factor concentration [%]
V simulation
VII simulation
X simulation
V measurement
VII measurement
X measurement

Figure 3. Validation Example. Warfarin acts via reduction of blood coagulation factor concentrations. The sensitivity of our blood coagulation model against blood coagulations factor deficiencies has been validated. The graph presents the predicted vs. experimental dependency of the INR on the relative factor concentration. (Experimental data taken from: Fisher Diagnostics: Thromboplastin Correlation Study 1999)

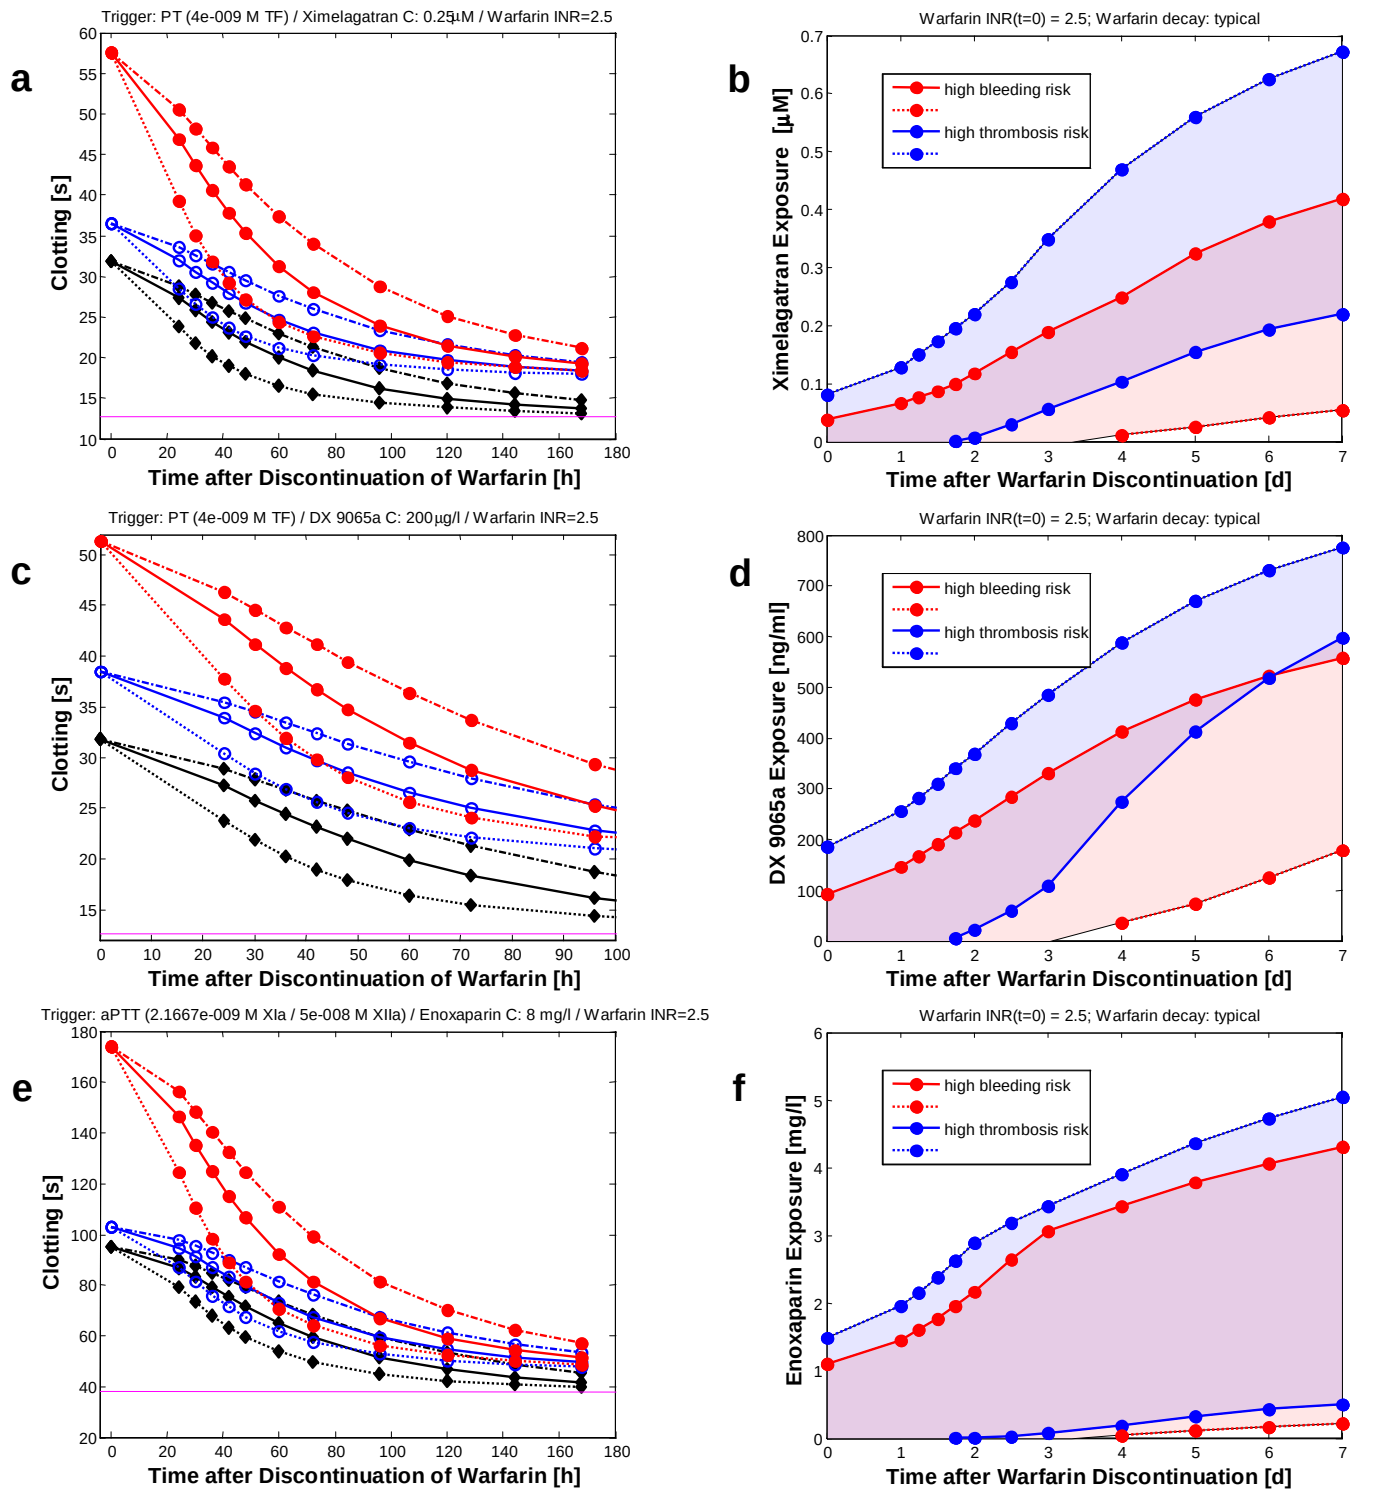
a
Trigger: PT (4e-009 M TF) / Ximelagatran C: 0.25µM / Warfarin INR=2.5
Clotting [s]
Time after Discontinuation of Warfarin [h]
b
Warfarin INR(t=0) = 2.5; Warfarin decay: typical
Ximelagatran Exposure [µM]
high bleeding risk
high thrombosis risk
Time after Warfarin Discontinuation [d]
c
Trigger: PT (4e-009 M TF) / DX 9065a C: 200µg/l / Warfarin INR=2.5
Clotting [s]
Time after Discontinuation of Warfarin [h]
d
Warfarin INR(t=0) = 2.5; Warfarin decay: typical
DX 9065a Exposure [ng/ml]
high bleeding risk
high thrombosis risk
Time after Warfarin Discontinuation [d]
e
Trigger: aPTT (2.1667e-009 M XIa / 5e-008 M XIIa) / Enoxaparin C: 8 mg/l / Warfarin INR=2.5
Clotting [s]
Time after Discontinuation of Warfarin [h]
f
Warfarin INR(t=0) = 2.5; Warfarin decay: typical
Enoxaparin Exposure [mg/l]
high bleeding risk
high thrombosis risk
Time after Warfarin Discontinuation [d]

Figure 4. Assessment of Synergistic Interaction and Derived Dosing Nomograms. The decay of Warfarin's impact on coagulation after its discontinuation (**a**, **c**, **e**, black lines; solid lines depict mean model, short dashed lines depict short Warfarin half-lives, long dashed lines represent long half-lives). Corresponding blue lines indicate joint impact of Warfarin residual action plus Ximelagatran, DX-9065a or Enoxaparin, assuming pure additivity. Red lines indicate the highly synergistic nature of interaction as predicted by the mechanistic model. Dosing nomograms were derived from INR-based guidelines for Warfarin alone (**b**, **d**, **f**). The broad dosing window found for Enoxaparin reflects the fact that aPTT is the more sensitive biomarker for this drug and INR-based considerations may therefore be too optimistic. Overall, our analysis suggests dose adjustment during the first few days of the new treatment.